Phage Therapy: Targeting Military Infectious Disease Threats

Merav Bassan, PhD meravb@biomx.com
Hila Sberro Livnat, PhD hilasl@biomx.com
Michael Billard mbillard@aphage.com

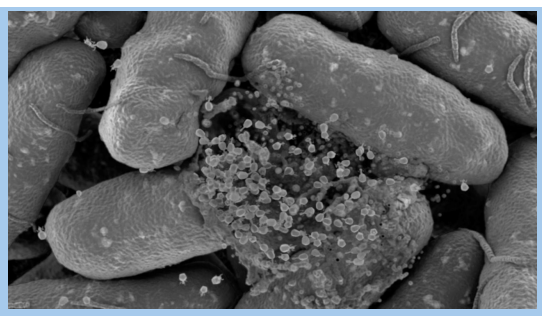

BiomX/APT: Who we are?
A leading, clinical-stage phage therapy company

- In March 2024, BiomX and Adaptive Phage Therapeutics (APT) merged
- The combined expertise and resources of both teams position the company as a **leading player in the phage therapy field**:
 - Two Phase 2 clinical programs expected to read out in 2025
 - ~80 compassionate use cases treated covering 13 infection types
 - Multiple active INDs in the US with indications granted Orphan Designation and Fast Track approvals by FDA along with other global regulatory approvals for studies
 - Significant government funding: >$40M received from Defense Health Agency, NIH
 - APT selected for Fierce Biotech’s 2023 Fierce 15 list
 - Non-traditional business
 - Investors: Deerfield, AMR Action Fund & Orbimed

What we do?
Exploit phage, a precise and natural solution to combat antibiotic-resistant infections

At this point, there are two ongoing clinical trials:

Personalized phage treatment approach

- Diabetic Foot Osteomyelitis (DFO) patients represent the majority of 160K lower limb amputations in diabetic patients annually in the US
- S. *aureus* is the most common pathogen in DFO
- Ongoing Phase 2b trial treating DFO patients with a personalized phage treatment

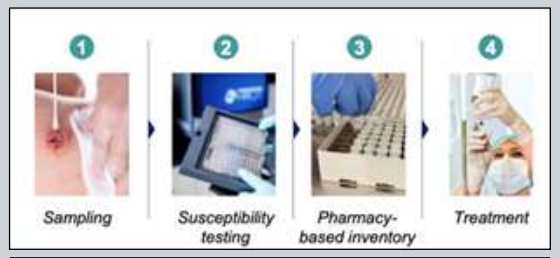
Sampling
Susceptibility testing
Pharmacy-based inventory
Treatment

Acknowledgement: The work is funded by the Naval Medical Research Command (NMRC) – Naval Advanced Medical Development (NAMD) program via the Medical Technology Enterprise Consortium (MTEC).

Fixed phage cocktail approach

- In CF patients, P. *aeruginosa* lung infections are a leading cause of morbidity and mortality; prolonged antibiotic treatments lead to significant resistance
- BX004, a phage cocktail product, was tested in a Phase 1b/2a study, showing promising results: 3 out of 21 (14.3%) patients in the BX004 arm converted to sputum culture negative for PsA after 10 days of treatment compared to 0 out of 10 (0%) in the placebo arm; signals of improvement in pulmonary function vs. placebo
- Phase 2b readout expected 3Q25

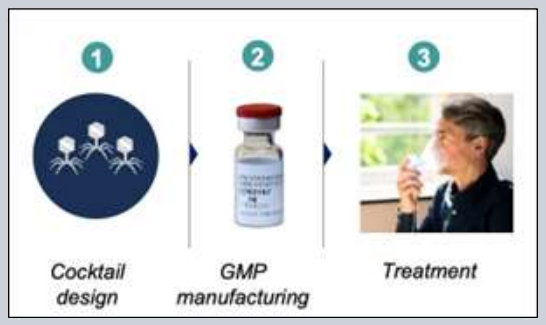
Cocktail design
GMP manufacturing
Treatment

What else do we bring to the table?
A platform that allows rapid development of phage-based therapies against species of interest

In addition to our current, ongoing clinical programs, we are well equipped and poised to rapidly develop new phage treatments against a variety of pathogens, including ESKAPE pathogens in a number of potential indications:

- A **large collections of both phage and clinically relevant bacteria:** hundreds of phage targeting multiple bacterial species, including ESKAPE and multi-drug resistant pathogens (e.g. MRSA, MSSA), as well as access to sources of clinical strains from different indications
- Robust **computational capabilities** that drive and accelerate our R&D at every stage; Thousands of bacterial and phage genomes sequenced and analyzed; multiple in-house tools to analyze genomes of phages and hosts
- **Synthetic biology capabilities:** We can tailor phages to deliver payloads of choice, creating a multitude of promising possibilities (Figure 1)
- **High Throughput Capabilities:** Evolution Experiments, Biofilm Assays, OD Automation
- With a strong interdisciplinary team, we are equipped to further harness phages **beyond traditional modalities** (e.g. for diagnostics and as a transfer vehicle)

Using a reporter gene as a payload to phage to create a rapid, potentially on-site, bacterial diagnostic testing

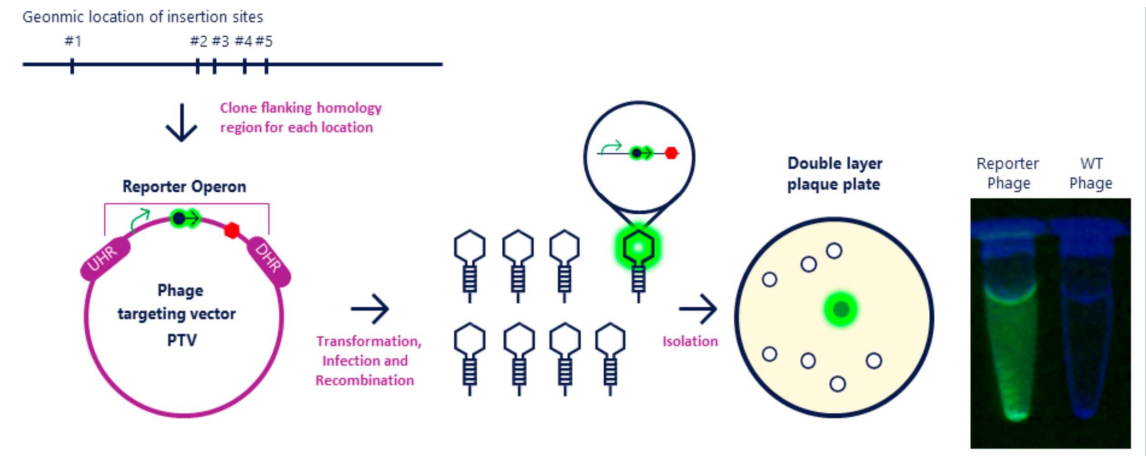
Geonmic location of insertion sites
#1
#2 #3 #4 #5
Clone flanking homology region for each location
Reporter Operon
UHR
DHR
Phage targeting vector PTV
Transformation, Infection and Recombination
Isolation
Double layer plaque plate
Reporter Phage
WT Phage

Figure 1. Rapid, potentially on-site, bacterial diagnostic testing using luminescent engineered phages is a culture- and isolation-free approach to detect the presence of phage-sensitive bacterial colonization in clinical samples. In this POC study (Zelbuch et al., 2021), we describe the engineering of two *K. pneumoniae* phages with the luciferase reporter gene (*nluc)*. We show that the reporter phages detect target bacteria directly on fecal samples with high sensitivity. This approach could be expanded to test presence of species of interest in different samples types (e.g. blood, saliva etc).

Example:
Development of a combined approach to precisely target *S. aureus* wound infections

The challenge we aim to address:

- Wound infections are frequent complications of combat casualties, characterized by multi-drug resistant organisms (MDROs), including MRSA and MSSA *S. aureus* (Tribble et al., 2019).
- The use of overly broad antibiotics has resulted in an increased risk of MDRO infection without improvement in long-term clinical outcomes
- The Joint Trauma System Clinical Practice Guidelines (CPGs) recommend avoiding unnecessary empiric use of broad-spectrum antibiotics and when available, to use a local antibiogram to guide empiric therapy. In addition, the current post-injury antimicrobial agent is Cefazolin, which is not effective against MRSA.

The approach:
1. Future diagnostics to enable accurate and rapid identification of infecting species

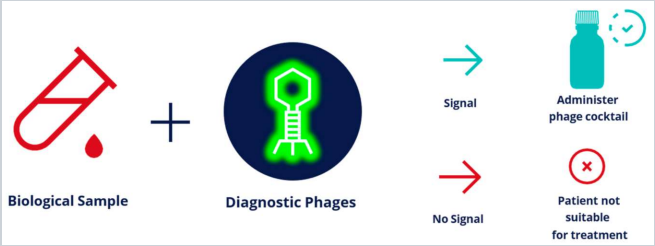
Biological Sample
Diagnostic Phages
Signal
Administer phage cocktail
No Signal
Patient not suitable for treatment

Mix of species-specific phages, each engineered with a different reporter protein to diagnose infecting species; a MRSA specific diagnostic tool

2. Treatment with an optimized cocktail

BiomX-APT is currently conducting a US DoD funded Phase 2b study investigating the safety, tolerability, and efficacy of personalized phage treatment for subjects with diabetic foot osteomyelitis due to *S. aureus*. **This trial will guide the development of a phage cocktail that will target *S. aureus* in combat-related wounds.** The data collected in this study will allow us to learn about:

1. Most efficient phage(s) in promoting complete healing of the *S. aureus* infected ulcers
2. Efficacy against biofilm, a significant challenge in wound care (Goswami et al., 2023)
3. Efficacy in combination with standard of care antibiotics
4. Efficacy against MSSA and MRSA strains

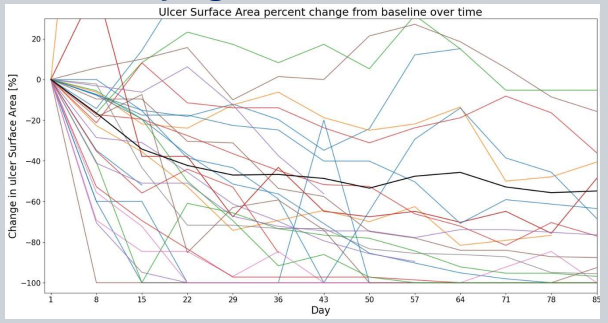
Ulcer Surface Area percent change from baseline over time
Change in ulcer Surface Area [%]
20
0
−20
−40
−60
−80
−100
Day
1
8
15
22
29
36
43
50
57
64
71
78
85

Reduction in ulcer area in response to treatment with different phages. This is an example of blinded data from the ongoing trial that will guide the development of an optimized cocktail.
Each color represents a different phage. Phage is selected per patient based on susceptibility of the patient’s strain to the phage.